977P

Interim results of phase 1 dose escalation study of YBL-006, a novel anti-PD-1 monoclonal antibody in advanced solid tumors

Keun-Wook Lee[1], John J. Park[2], Do-Youn Oh[3], Se Hyun Kim[1], Dhanusha Sabanathan[2], Tae Min Kim[3], Myungsuk Kim[4], Jaebong Yoon[4], Han Seung Lee[4], Seongyeol Park[5], Kyunghyun Paeng[6], Chan-Young Ock[6]

[1]Seoul National University Bundang Hospital, Seoul National University College of Medicine, Seongnam, Korea; [2]Department of Clinical Medicine, Macquarie University, North Ryde, NSW, Australia; [3]Seoul National University Hospital, Cancer Research Institute, Seoul National University College of Medicine, Seoul, Korea; [4]Y-Biologics Inc., Daejeon, Republic of Korea; [5]Genome Insight Inc., Daejeon, Republic of Korea; [6]Lunit Inc., Seoul, Republic of Korea

Background

- PD-1 (programmed cell death protein 1) is an immune check-point receptor, which is widely expressed on immune cells. Binding of PD-1 to its ligand PD-L1 can trigger an inhibitory signal, leading to reduced T-cell proliferation and anti-tumor immunity.[1)]
- YBL-006 is a fully human anti-PD-1 mAb, which was derived from a human B cell cDNA antibody library using phage display technology.
- YBL-006 has a wider PD-1-binding interface (**Figure 1**) and shows higher affinity to PD-1 compared to nivolumab and pembrolizumab.[2)]

Objectives

The objectives of the dose escalation cohort in this phase 1 open label study were to:

- characterize safety, tolerability and PK profile of YBL-006
- evaluate preliminary efficacy, immunogenicity and PD-1 receptor occupancy of YBL-006

Methods

Study design: Phase 1 dose escalation study in patients with advanced solid tumor (NCT04450901)

- Total of 11 patients across 4 dose escalation cohorts A1 to A4 were administered YBL-006 as a 60-minute IV infusion on Day 1 and Day 15 of every 28-day cycle.
- As a patient has not experienced a dose-limiting toxicity (DLT) during 28-day DLT evaluation interval at the starting dose of 0.5 mpk (mg/kg, cohort A1), all subsequent dosing cohorts followed traditional 3 + 3 dose escalation criteria **(Figure 2)**.
- Tumor responses were assessed every 8 weeks according to RECIST v1.1.
- Adverse events (AEs) were graded using the CTCAE v5.
- Exploratory biomarker analyses included;
 - Whole exome sequencing to assess tumor mutational burden (TMB)
 - Lunit SCOPE IO[3)] to assess the density of intra-tumoral tumor-infiltrating lymphocyte (TIL)
- Dose expansion cohort will open to patients with advanced solid tumors including special interest tumor types administering fixed doses of YBL-006, and a recommended phase 2 dose (RP2D) will be established.

Figure 1. Comparison of YBL-006 binding interface to PD-1 with other anti-PD-1 mAbs

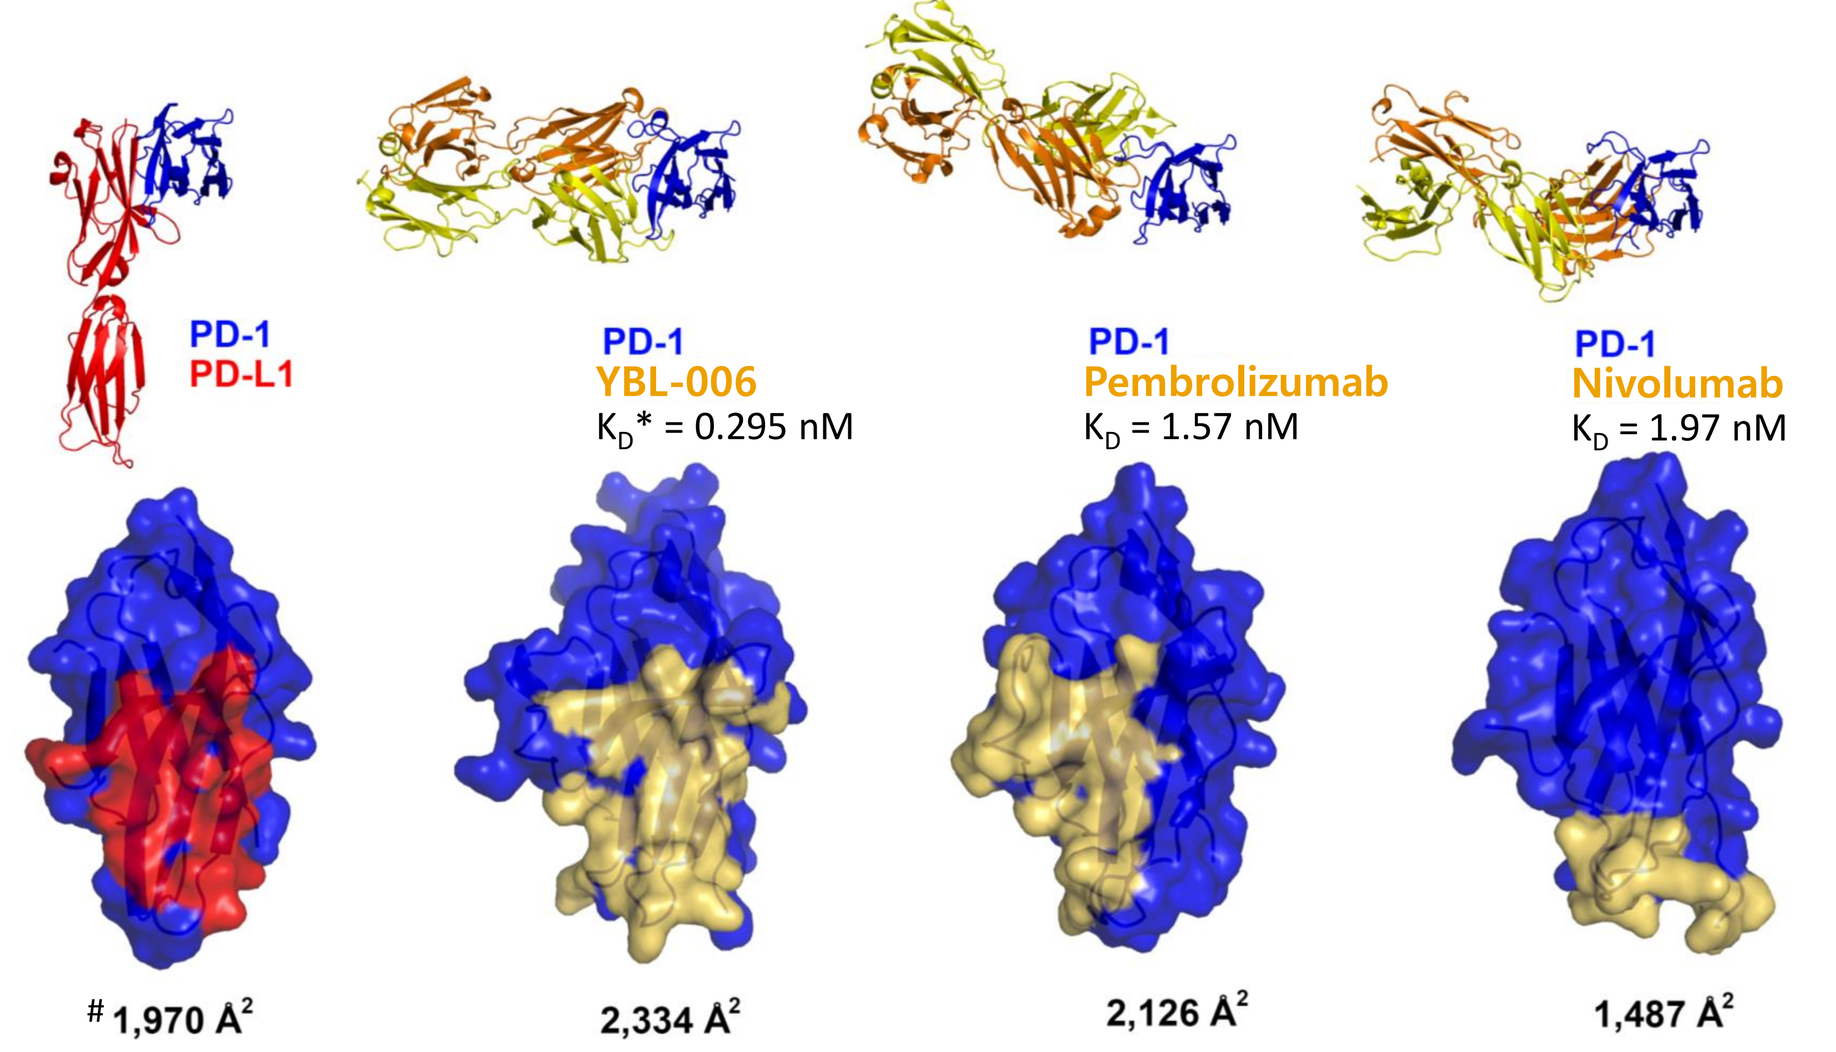

PD-1
PD-L1
PD-1
YBL-006
K_D* = 0.295 nM
PD-1
Pembrolizumab
K_D = 1.57 nM
PD-1
Nivolumab
K_D = 1.97 nM
1,970 Å²
2,334 Å²
2,126 Å²
1,487 Å²

* Binding affinity measured by SPR.
\# Buried surface area

Figure 2. Overall Study design

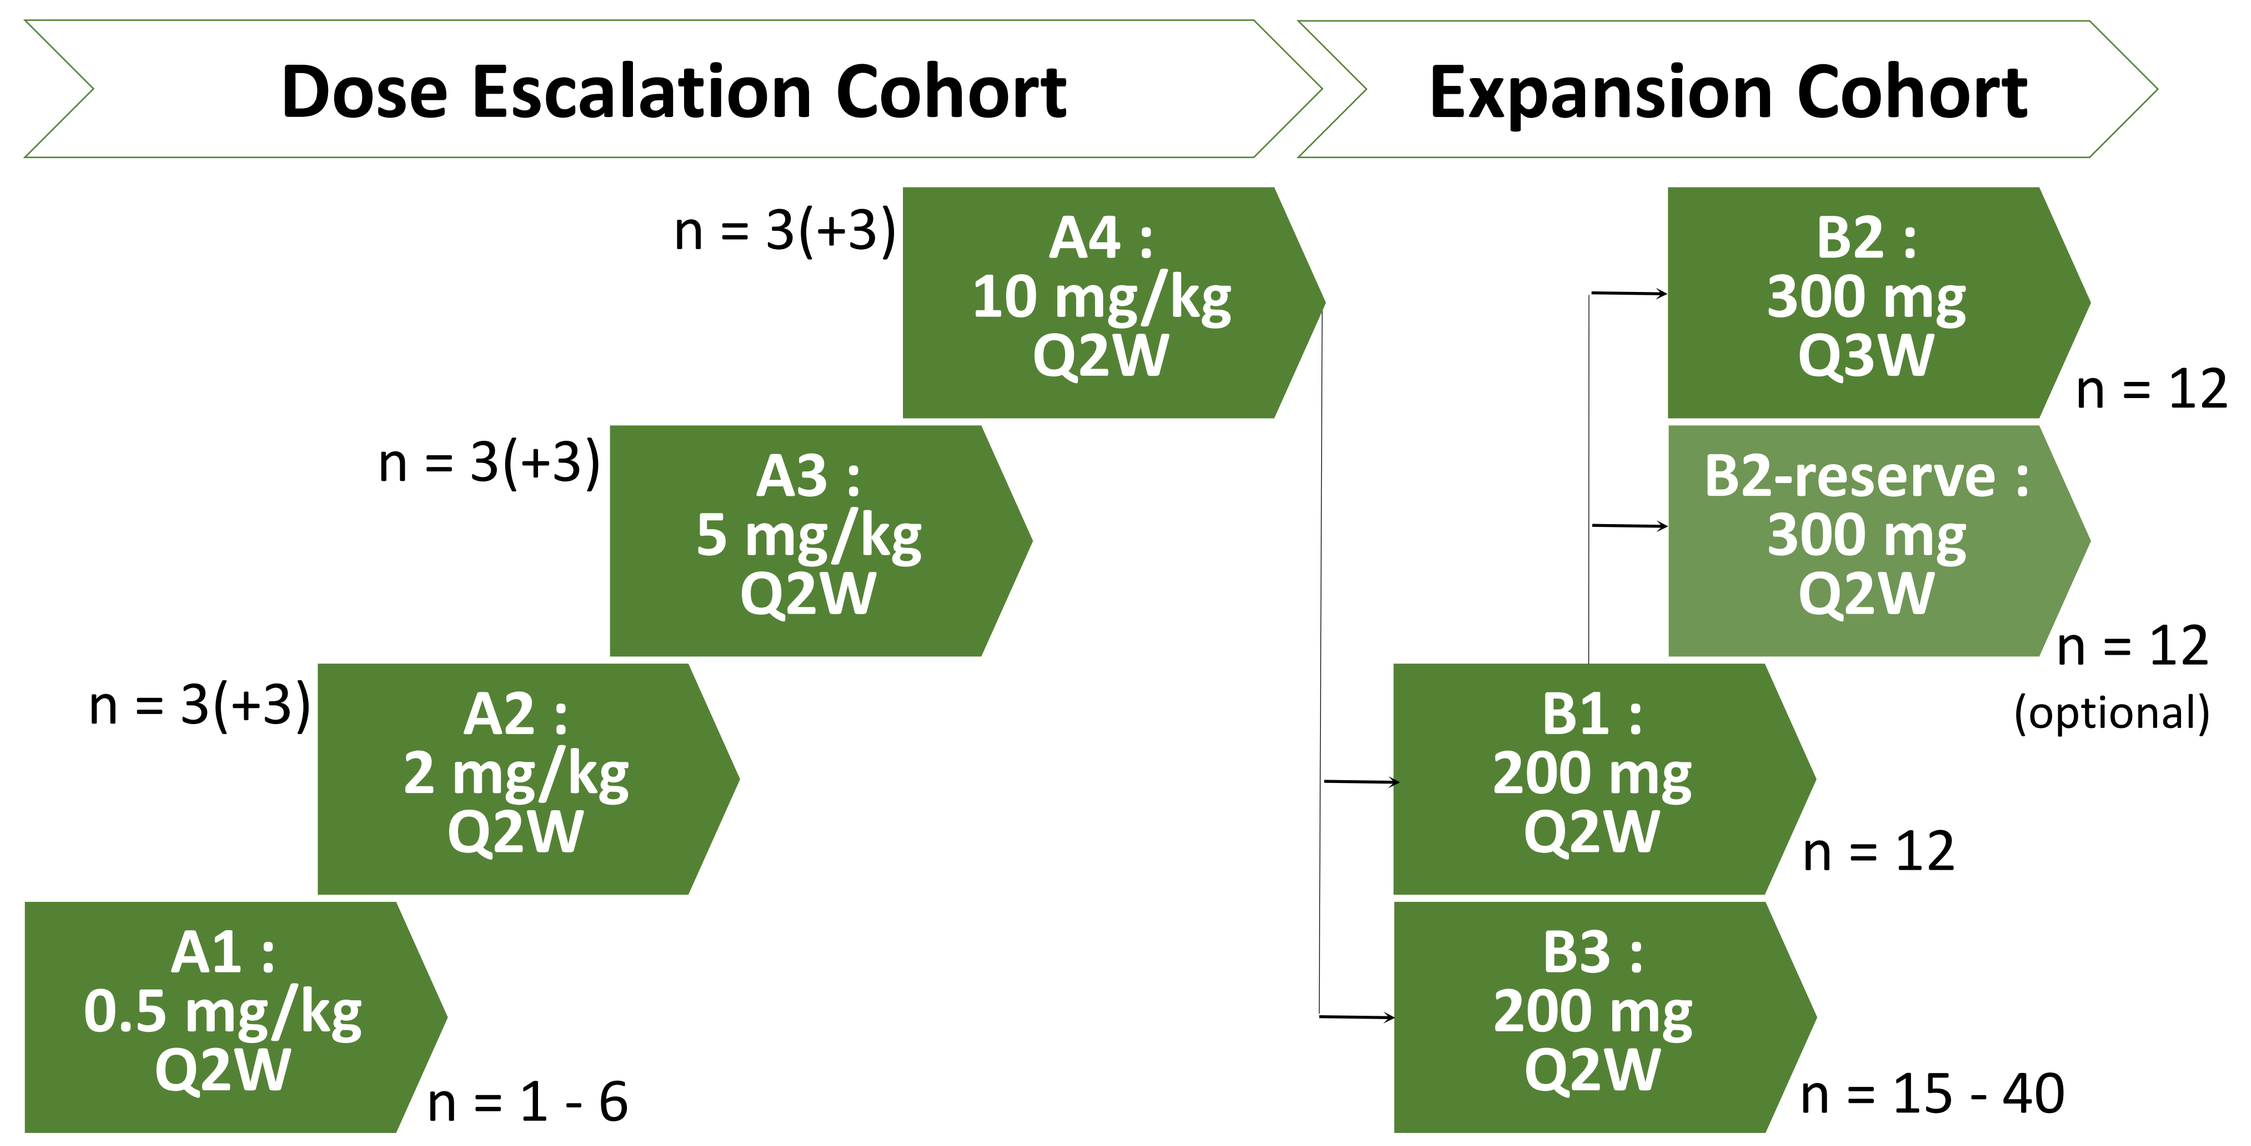

Dose Escalation Cohort
Expansion Cohort
n = 3(+3)
A4 : 10 mg/kg Q2W
n = 3(+3)
A3 : 5 mg/kg Q2W
n = 3(+3)
A2 : 2 mg/kg Q2W
A1 : 0.5 mg/kg Q2W
n = 1 - 6
B2 : 300 mg Q3W
n = 12
B2-reserve : 300 mg Q2W
n = 12 (optional)
B1 : 200 mg Q2W
n = 12
B3 : 200 mg Q2W
n = 15 - 40

Results

As of April 27, 2021 (data cut-off date), total of 11 patients (median age, 62 years) with advanced solid tumors were enrolled in the dose escalation cohort.

Patient characteristics

Patient characteristics and exposure to YBL-006 are summarized in **Table 1.**

Table 1. Patient baseline characteristics

	0.5 mpk (n = 1)	2 mpk (n = 3)	5 mpk (n = 4)	10 mpk (n = 3)	Total (n = 11)
Age, years					
median (range)	76 (76)	65 (52 - 65)	55.5 (44 - 72)	57 (22 - 68)	62 (22 - 76)
≥ 65 years, n (%)	1 (100.0)	2 (66.7)	1 (25.0)	1 (33.3)	5 (45.5)
Gender, n (%)					
Male	1 (100.0)	3 (100.0)	2 (50.0)	1 (33.3)	7 (63.6)
Female	0	0	2 (50.0)	2 (66.7)	4 (36.4)
ECOG performance status score, n (%)					
0	0	3 (100.0)	0	2 (66.7)	5 (45.5)
1	1 (100.0)	0	4 (100.0)	1 (33.3)	6 (54.5)
Cancer type, n (%)					
Colorectal cancer	0	0	2**(50.0)	0	2 (18.1)
Sarcoma	1 (100.0)	0	0	2 (66.7)	3 (27.3)
Squamous cell carcinoma	0	2*(66.7)	1***(25.0)	0	3 (27.3)
Nasopharyngeal carcinoma	0	1 (33.3)	0	0	1 (9.1)
Thymic sarcomatoid carcinoma	0	0	1 (25.0)	0	1 (9.1)
Neuroendocrine carcinoma	0	0	0	1 (33.3)	1 (9.1)
Prior chemotherapy					
Line of therapy, median (range)	1 (1)	1 (0 - 6)	2 (1 - 5)	2 (1 - 5)	1 (0 - 6)

* Penile (n = 1), Anal (n = 1)
** MSI-H (n = 1), MSS (n = 1)
*** Anal (n = 1)

Pharmacokinetic and pharmacodynamic analysis

- YBL-006 showed a linear PK prolife with $T_{1/2}$ of approximately 8 days **(Figure 3 and Table 2)**.
- Both PD-1 receptor occupancy (PD-1 RO) and serum IFN-γ level were increased by 3 - 4 times after the first dosing.
 - PD-1 RO was 64.0% (95% CI, 46.8 - 81.2) at 2 weeks and 68.4% (95% CI, 53.6 - 83.3) at 4 weeks after administration of YBL-006 compared with 19.0% (95% CI, 16.5 - 21.5) before administration **(Figure 4A)**.
 - Serum IFN-γ levels were 2.65 ng/mL (95% CI, 1.27 - 4.03) at 8 hours after administration of YBL-006 compared with 0.70 ng/mL (95% CI, 0.36 - 1.04) before administration **(Figure 4B)**.

Figure 3. Pharmacokinetic (PK) profiles of YBL-006

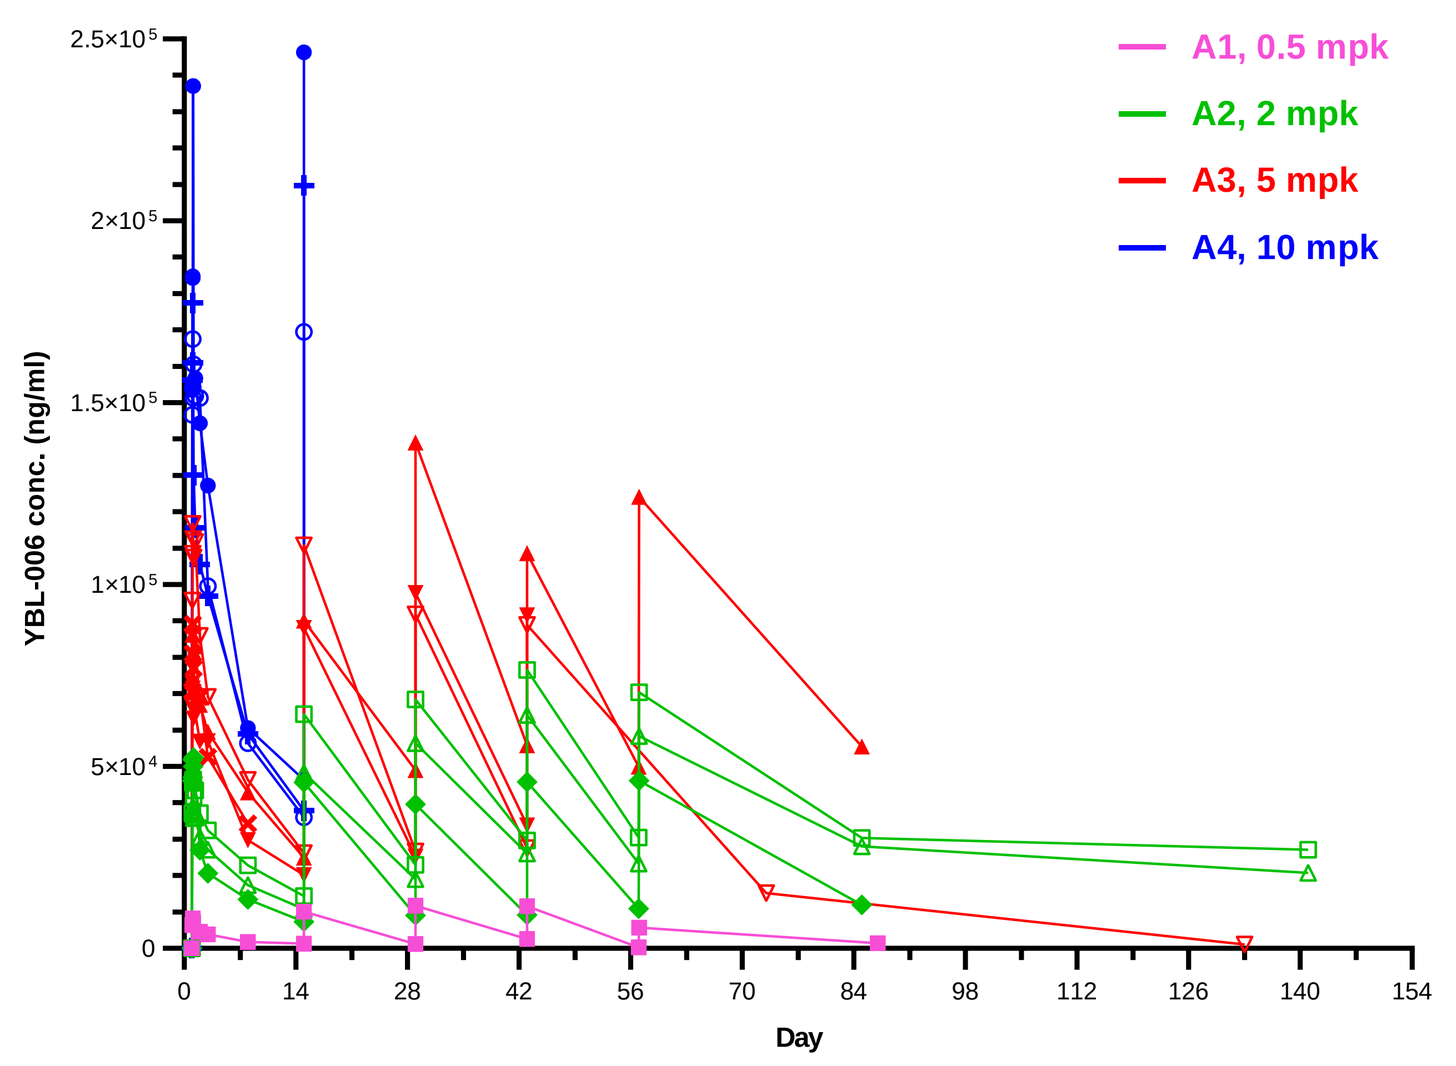

A1, 0.5 mpk
A2, 2 mpk
A3, 5 mpk
A4, 10 mpk
YBL-006 conc. (ng/ml)
Day

Table 2. Pharmacokinetic (PK) parameters

	0.5 mpk (n = 1)	2 mpk (n = 3)	5 mpk (n = 4)	10 mpk (n = 3)
$T_{1/2}$ (days)	7.48	8.68 (8.08 - 9.39)	8.26 (6.18 - 9.64)	8.45 (7.88 - 9.11)
C_{max} (μg/mL)	8.22	47.1 (46.5 - 52.5)	88.9 (77.3 - 116.7)	177.5 (167.5 - 237.1)
$AUC_{0\text{-}last}$ (μg/mL·hr)	851	6,465 (5,486 - 7,757)	13,360 (8,214 - 17,140)	24,550 (22,280 - 28,040)

median (range)

Figure 4. PD-1 receptor occupancy and IFN-γ level

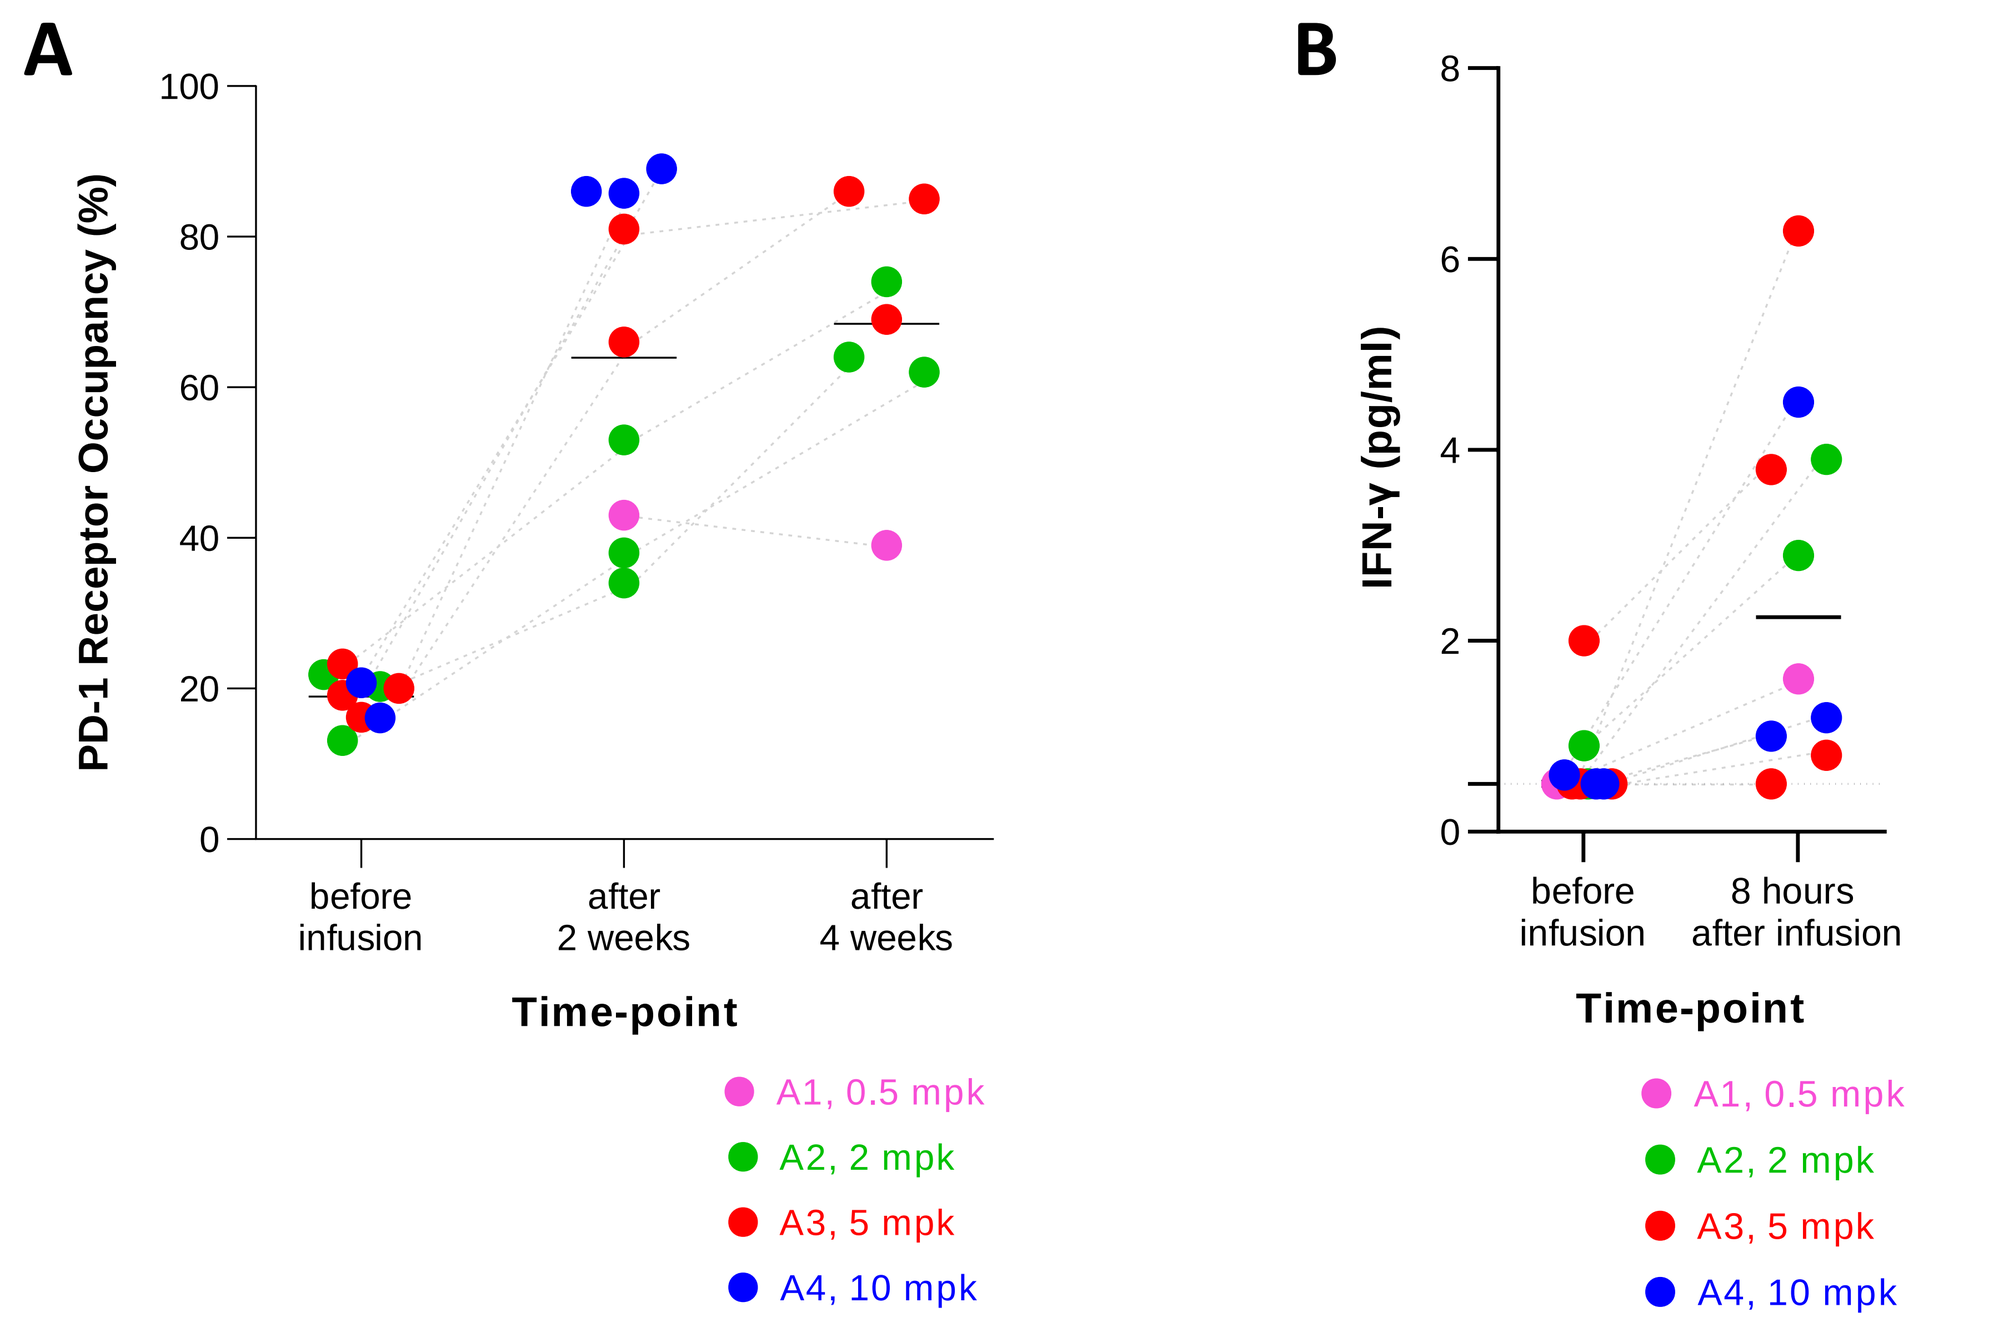

A
PD-1 Receptor Occupancy (%)
before infusion
after 2 weeks
after 4 weeks
Time-point
B
IFN-γ (pg/ml)
before infusion
8 hours after infusion
Time-point
A1, 0.5 mpk
A2, 2 mpk
A3, 5 mpk
A4, 10 mpk

Tumor response

Total of 10 patients were available for tumor response evaluation. The best overall responses included one complete response (penile squamous cell carcinoma, 2 mpk) and one partial response (anal squamous cell carcinoma, 2 mpk) cases with durable responses lasting more than 30 and 14 weeks, respectively, and two stable disease cases were also observed **(Figure 5, 6 and Table 3)**.

Figure 5. Tumor responses to YBL-006 (n = 10)

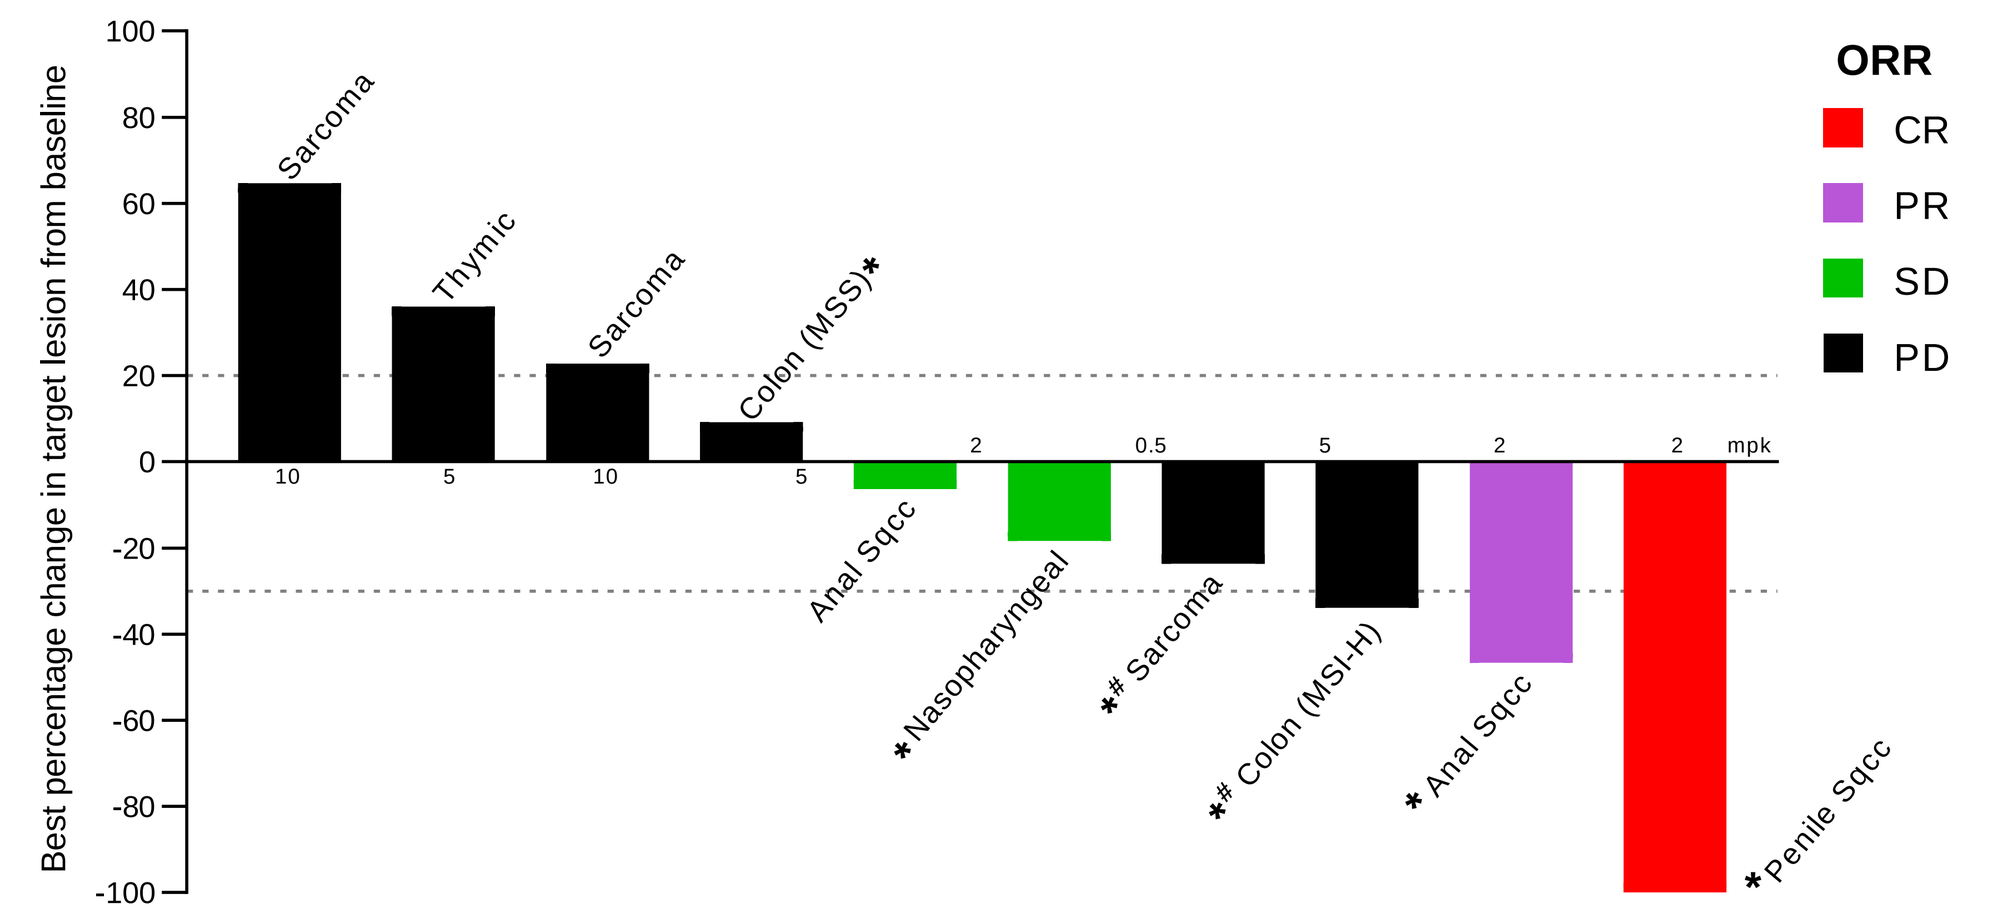

Best percentage change in target lesion from baseline
Sarcoma
Thymic
Sarcoma
Colon (MSS)*
Anal Sqcc
* Nasopharyngeal
*# Sarcoma
*# Colon (MSI-H)
* Anal Sqcc
* Penile Sqcc
mpk
ORR
CR
PR
SD
PD

* Biomarker analysis
\# Target lesions were assessed as SD and PR for sarcoma and colon (MSI-H) cancer patients, respectively, however their overall responses were assessed as PD since the new lesions were found in these patients.

Figure 6. Changes in target lesion over time (n = 10)

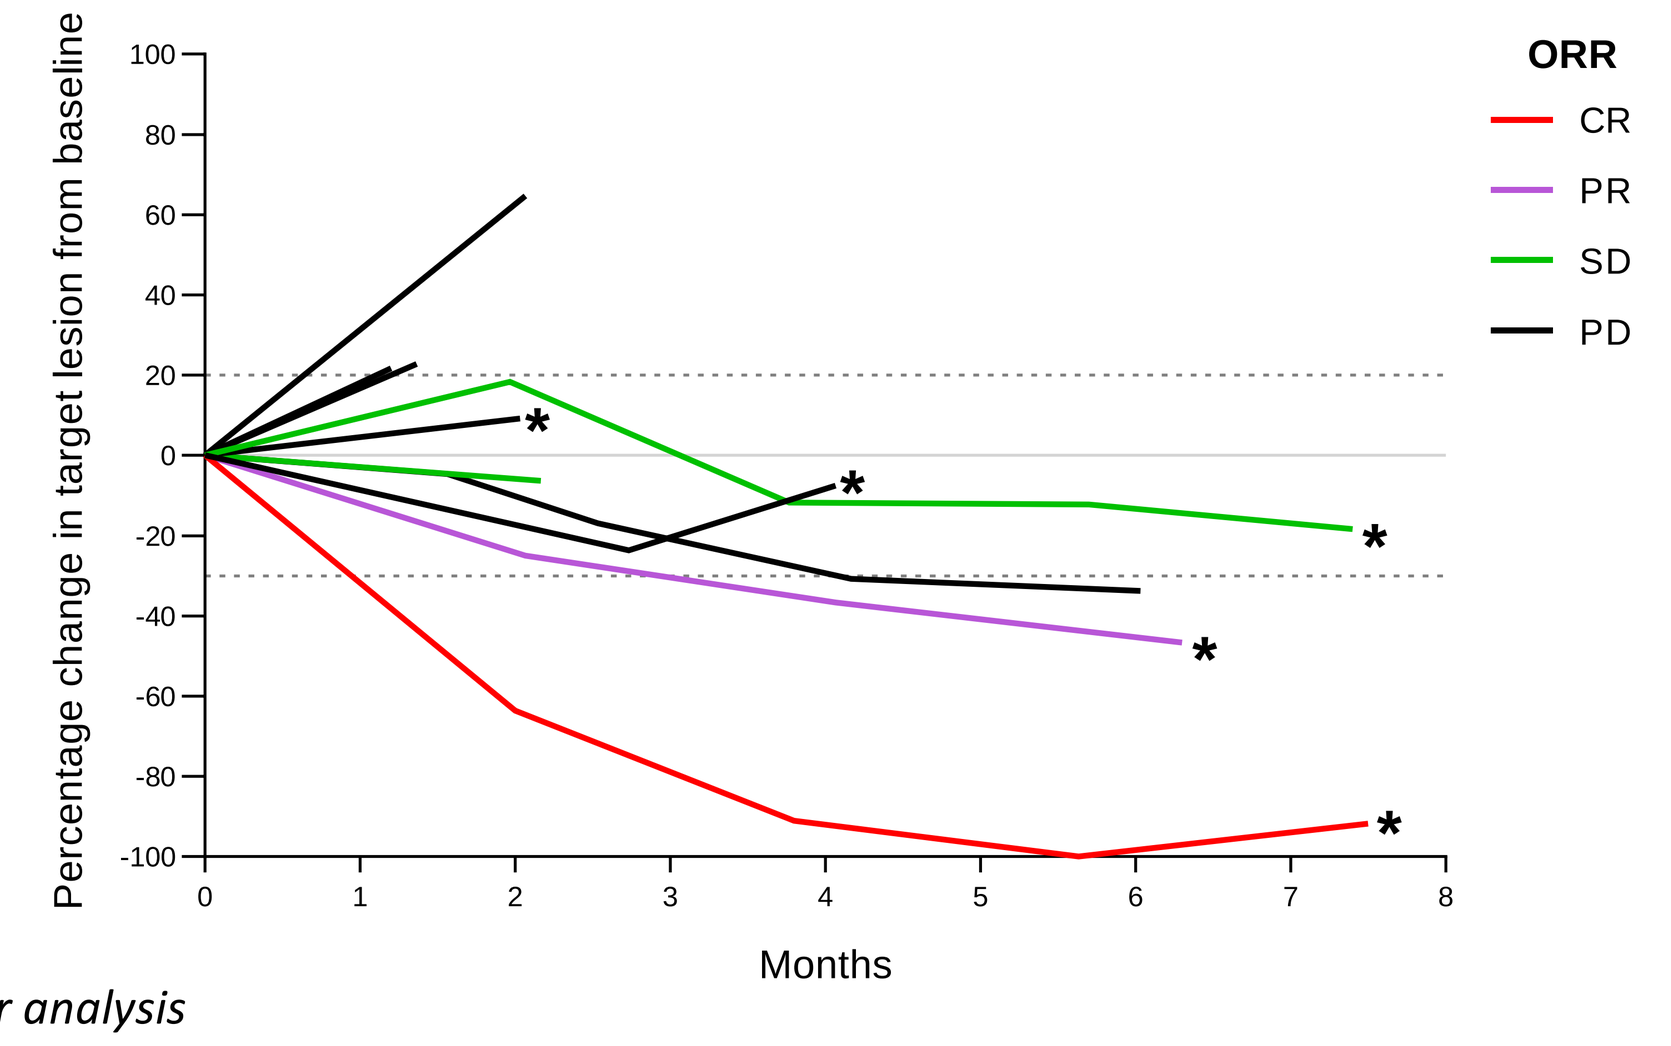

Percentage change in target lesion from baseline
Months
ORR
CR
PR
SD
PD

* Biomarker analysis

Table 3. Tumor response by investigator assessment (n = 10)

	0.5 mpk (n = 1)	2 mpk (n = 3)	5 mpk (n = 4)	10 mpk (n = 2)	Total (n = 10)
Best overall response, n (%)					
Complete response	0	1 (33.3)	0	0	1 (10.0)
Partial response	0	1 (33.3)	0	0	1 (10.0)
Stable disease	0	1 (33.3)	1 (25.0)	0	2 (20.0)
Progressive disease	1 (100.0)	0	3 (75.0)	2 (100.0)	6 (60.0)
Overall response rate*, %	0	66.7	0	0	20
Clinical benefit rate, %**	0	100.0	25.0	0	40
Treatment duration, weeks median (range)	15 (15)	32.5 (29 - 36)	6 (0 - 19)	4 (2 - 6)	17 (0 - 36)
Time to response, weeks median (range)	5 (5)	24.5 (20 - 29)	4.5 (0 - 9)	0*** (0)	14.5 (0 - 29)
Duration of overall response, weeks	0 (0)	16.3 (0 - 29)	0 (0)	0 (0)	16.3 (0 - 29)

* Overall response rate = {CR (n)+PR (n)}/Evaluable patients (n = 10)·100
** Clinical benefit rate = {CR (n)+PR (n)+SD (n)}/Evaluable patients (n = 10)·100
*** The last visit date = Response assessment date

Safety

- TEAEs (excluded 'Not related') of any grade were reported in **Table 4.**
- No dose limiting toxicities (DLTs) or deaths related to YBL-006 have been reported.
- The most common AEs of ≥ Grade 2 related to YBL-006 were rash (36.4%), fatigue (18.2%), fever (18.2%) and hypothyroidism (9.1%).

Table 4. Treatment-emergent adverse events (TEAEs)*

TEAEs of Grade 1 or 2, or ≥ Grade 3 in any patient	Number of patient (%**) Grade 1 or 2	≥ Grade 3
Rash***	4 (36.4)	0
Fatigue	2 (18.2)	0
Fever	2 (18.2)	0
Cytokine release syndrome****	1 (9.1)	0
Elevated CK	1 (9.1)	0
Flu-like symptom	1 (9.1)	0
Hypothyroidism	1 (9.1)	0
Hypertension	1 (9.1)	0
Intermittent vomiting	1 (9.1)	0
Irritated eyes	1 (9.1)	0
Lichenoid (oral)	1 (9.1)	0
Nausea	1 (9.1)	0
Oral candidiasis	1 (9.1)	0
Oral thrush	1 (9.1)	0
Pain of shoulder	1 (9.1)	0
Urticaria	1 (9.1)	0

* Excluded 'Not related' AEs with study treatment
** % = Number of patient per each AE (n)/total number of patient (n=11)·100
*** 1 Pruritic rash, 1 Maculopapular rash, 1 Pruritus and 1 Skin pruritis cases (Grade 1 or 2)
**** Serious TEAE

Biomarker analysis

Tumor samples of 2 responders (CR and PR) harbored high level of TMB (8.3 and 9.3 per megabase) and intra-tumoral TIL density (66.1% and 95.8%) **(Table 5 and Figure 7)**.

Table 5. Exploratory biomarker analysis (n = 5)

ID	Dose (mg/kg)	Cancer type	Whole exome Sequencing: TMB	Whole exome Sequencing: Driver mutation	Lunit SCOPE IO: Inflammed	Lunit SCOPE IO: Immune-Excluded	Lunit SCOPE IO: Immune-Desert	Best overall response	Target lesion %	PFS1 (weeks)	PFS2 (weeks)
112-001	2	Penile squamous cell carcinoma	8.3 / Mb	PIK3CA E545K STK11 Q100X (LOH)	66.1%	26.2%	7.60%	CR	-100%	36	N/A
121-002	2	Anal squamous cell carcinoma	9.3 / Mb	PIK3CA E545K JAK2/CD274 co-amplification	95.8%	3.6%	0.60%	PR	-46.6%	36	N/A
112-002	2	Nasopharyngeal carcinoma		*QC fail*	77.1%	11.4%	11.40%	SD	-18.3%	31	N/A
121-001	0.5	Undifferentiated Pleomorphic Sarcoma	1.7 / Mb	PDGFRA amplification ARID1B D1772N		*QC fail*		PD	-23.7%	10	5
112-004	5	MSS Colon cancer	5.5 / Mb	CDX2 amplification CCND3 amplification	25.1%	51.0%	23.90%	PD	9.5%	8	N/A

Figure 7. Response to YBL-006 in patient ID #121-002

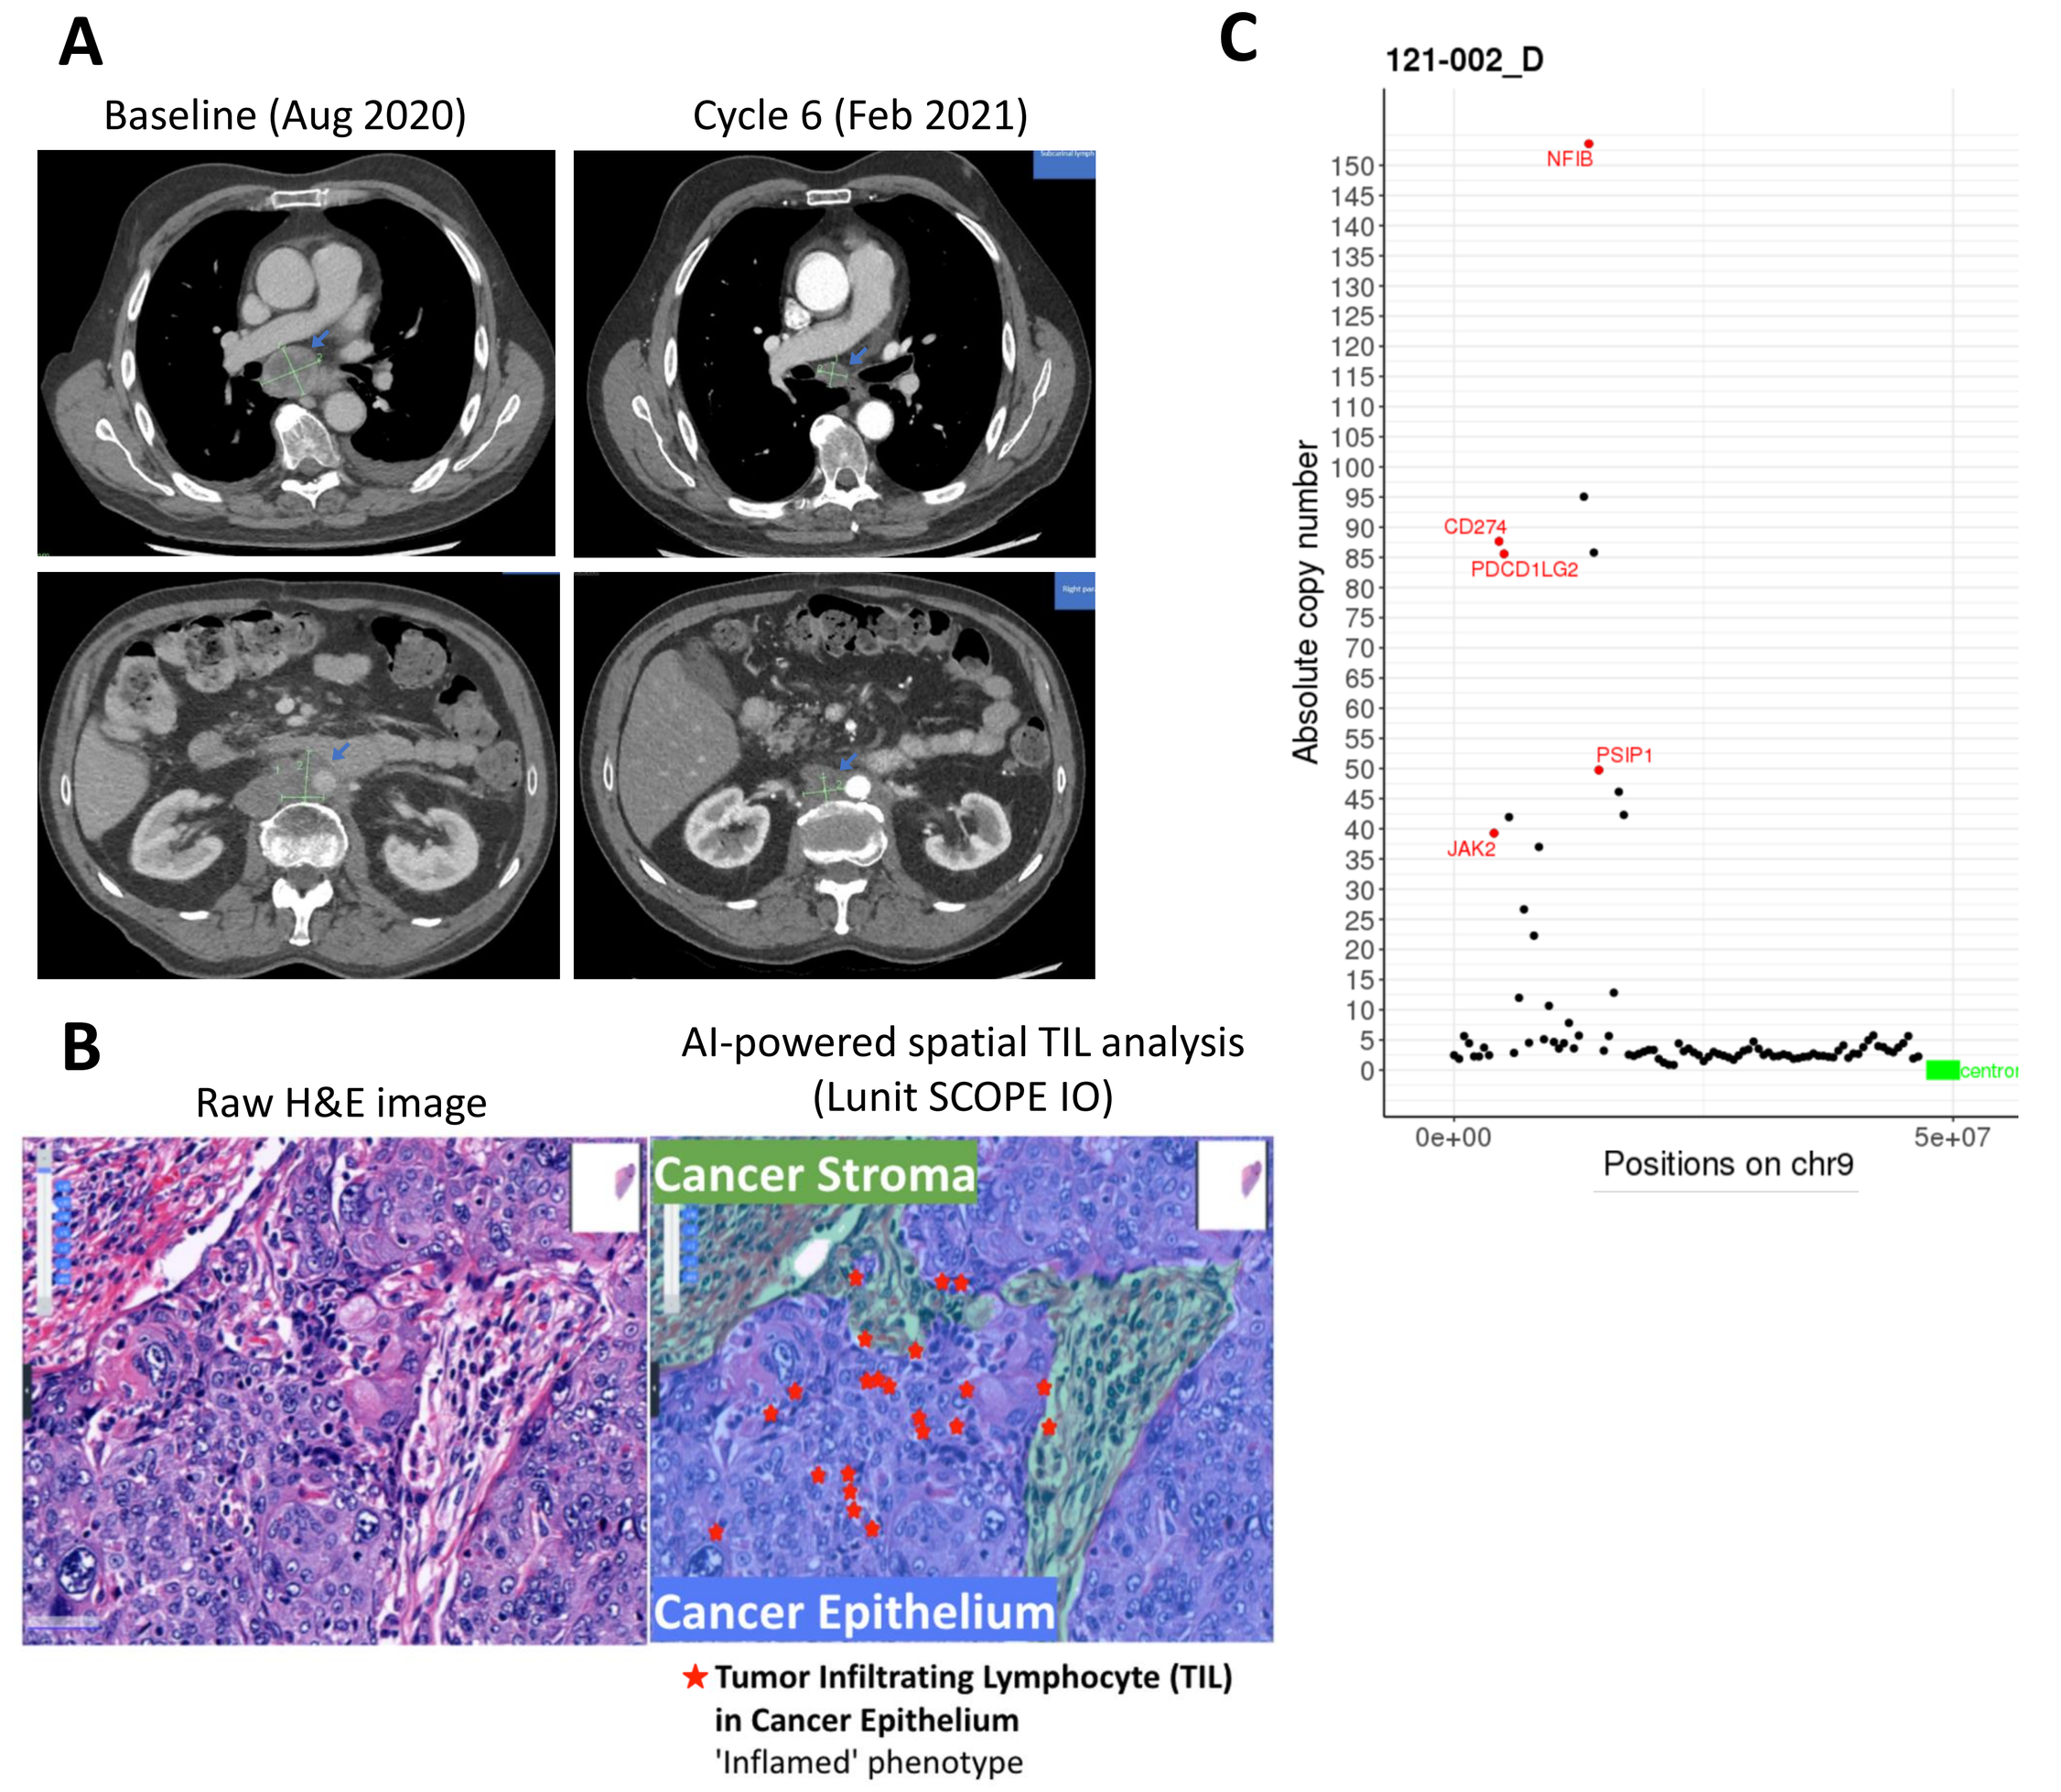

A
Baseline (Aug 2020)
Cycle 6 (Feb 2021)
C
121-002_D
NFIB
CD274
PDCD1LG2
PSIP1
JAK2
Absolute copy number
Positions on chr9
B
Raw H&E image
AI-powered spatial TIL analysis (Lunit SCOPE IO)
Cancer Stroma
Cancer Epithelium
Tumor Infiltrating Lymphocyte (TIL) in Cancer Epithelium
'Inflamed' phenotype

A. CT scan of the abdomen in patient ID #121-002
Case study #121-002 : *# 66/M # anal squamous cell carcinoma, stage IV (2015)*
s/p mitomycin + 5-FU, palliative radiotherapy, and metastatectomy (2015 - 2016) → PD (2020)
started YBL-006 phase I study (2 mpk, Q2W)
experienced G1 cytokine release syndrome at C1D12 → recovered with best supportive care
RECIST v1.1: Baseline → Cycle 2 (SD, -25%) → Cycle 4 (PR, -36.6%) → Cycle 6 (PR, -46.6%)
still under YBL-006 treatment without intolerability (PFS 36 weeks, DoR 29 weeks)
B. Lunit SCOPE IO (AI-TIL analyzer for H&E specimen): High TIL in cancer epithelium (inflamed phenotype)
C. JAK2 & CD274 (PD-L1): co-amplification on chromosome 9p24

Conclusions

- YBL-006 was well tolerated in the studied dose range and showed anti-tumor activity in patients with advanced solid tumors.
- YBL-006 follows a linear pharmacokinetics with $T_{1/2}$ of approximately 8 days.
- PD-1 RO and IFN-γ release were increased by 3 - 4 times after administration of YBL-006.
- The trial is open for dose expansion cohort using fixed doses. Clinical trial information: NCT04450901.

Disclosure

Dr. Keun-Wook Lee *grants from Y-Biologics (to his institution for conducting clinical trials)*

References

1) Y Chen , et al. Front Immunol. 2020 May 29;11:1088
2) J Yoon, et al. Poster presentation(#3367) at AACR Annual Meeting 2020
3) J Shen, et al. Journal of Clinical Oncology. 2021 May 28;39(15):2607-2607

Acknowledgments

This study was sponsored by Y-Biologics Inc.
For any questions regarding this poster presentation, please contact mskim@ybiologics.com

Presented at the (ESMO) Congress, e-Poster section, September 16-21th, 2021.